388P

Early tumor shrinkage and depth of response analyses in metastatic colorectal cancer treated with first-line mFOLFOX6 plus panitumumab or bevacizumab: Results from the phase 3 PARADIGM trial

Kei Muro,[1] Jun Watanabe,[2] Kohei Shitara,[3] Kentaro Yamazaki,[4] Hisatsugu Ohori,[5] Manabu Shiozawa,[6] Hirofumi Yasui,[4] Eiji Oki,[7] Takeo Sato,[8] Takeshi Naitoh,[9] Yoshito Komatsu,[10] Takeshi Kato,[11] Masamitsu Hihara,[12] Junpei Soeda,[12] Kouji Yamamoto,[13] Kiwamu Akagi,[14] Atsushi Ochiai,[15] Hiroyuki Uetake,[16] Katsuya Tsuchihara,[17] Takayuki Yoshino[18]

[1]Department of Clinical Oncology, Aichi Cancer Center Hospital, Nagoya, Japan; [2]Gastroenterological Center, Yokohama City University Medical Center, Yokohama, Japan; [3]Department of Gastroenterology and Gastrointestinal Oncology, National Cancer Center Hospital East, Chiba, Japan; [4]Division of Gastrointestinal Oncology, Shizuoka Cancer Center, Shizuoka, Japan; [5]Division of Medical Oncology, Japanese Red Cross Ishinomaki Hospital, Miyagi, Japan; [6]Division of Gastrointestinal Surgery, Kanagawa Cancer Center, Kanagawa, Japan; [7]Department of Surgery and Science, Graduate School of Medical Sciences, Kyushu University, Fukuoka, Japan; [8]Research and Development Center for Medical Education, Department of Clinical Skills Education, Kitasato University School of Medicine, Sagamihara, Japan; [9]Department of Lower Gastrointestinal Surgery, Kitasato University School of Medicine, Sagamihara, Japan; [10]Division of Cancer Chemotherapy, Hokkaido University Hospital Cancer Center, Sapporo, Japan; [11]Department of Surgery, National Hospital Organization Osaka National Hospital, Osaka, Japan; [12]Japan Medical Affairs, Japan Oncology Business Unit, Takeda Pharmaceutical Company Ltd., Tokyo, Japan; [13]Department of Biostatistics, Yokohama City University School of Medicine, Yokohama, Japan; [14]Division of Molecular Diagnosis and Cancer Prevention, Saitama Cancer Center, Saitama, Japan; [15]Pathology Division, Exploratory Oncology Research & Clinical Trial Center, National Cancer Center, Chiba, Japan; [16]National Hospital Organization, Disaster Medical Center, Tokyo, Japan; [17]Division of Translational Informatics, Exploratory Oncology Research & Clinical Trial Center, National Cancer Center, Chiba, Japan; [18]Department of Gastroenterology and Gastrointestinal Oncology, National Cancer Center Hospital East, Kashiwa, Japan

Introduction

- The primary analysis of the PARADIGM trial (NCT02394795) demonstrated that panitumumab plus modified FOLFOX6 (mFOLFOX6) significantly improved overall survival (OS) compared with bevacizumab plus mFOLFOX6 in patients with *RAS* wild-type (WT) and left-sided metastatic colorectal cancer (mCRC) **(Table 1)**[1]

Table 1: Primary efficacy outcomes from the phase 3 PARADIGM trial

Parameter	Panitumumab + mFOLFOX6	Bevacizumab + mFOLFOX6	HR (95% CI)
Left-sided mCRC	n=312	n=292	
Median OS, mo (95% CI)	37.9 (34.1–42.6)	34.3 (30.9–40.3)	0.82 (0.68–0.99); *P*=0.031
Median PFS, mo (95% CI)	13.7 (12.7–15.3)	13.2 (11.4–14.5)	0.98 (0.82–1.17)
Response rate, % (95% CI)	80.2 (75.3–84.5)	68.6 (62.9–74.0)	NA
Right-sided mCRC	n=84	n=103	
Median OS, mo (95% CI)	20.2 (15.2–32.0)	23.2 (18.5–29.1)	1.09 (0.79–1.51); *P*=0.605
Median PFS, mo (95% CI)	7.7 (6.3–10.6)	10.6 (7.6–13.0)	1.23 (0.91–1.67)
Response rate, % (95% CI)	54.9 (43.5–65.9)	63.1 (53.0–72.4)	NA
Overall population	n=400	n=402	
Median OS, mo (95% CI)	36.2 (32.0–39.0)	31.3 (29.3–34.1)	0.84 (0.72–0.98); *P*=0.030
Median PFS, mo (95% CI)	12.9 (11.3–13.6)	12.0 (11.3–13.5)	1.01 (0.87–1.18)
Response rate, % (95% CI)	74.9 (70.3–79.1)	67.3 (62.4–71.9)	NA

- Early tumor shrinkage (ETS) and depth of response (DpR) are on-treatment prognostic factors for favorable OS and progression-free survival (PFS) outcomes associated with chemotherapy for mCRC[2,3]
- Here, we report ETS and DpR from the PARADIGM trial and the correlation of these prognostic factors with OS and PFS
- The objective was to examine the correlation of ETS with OS and PFS

Methods

Figure 1: PARADIGM: Phase 3, randomized, open-label, multicenter study (NCT02394795)

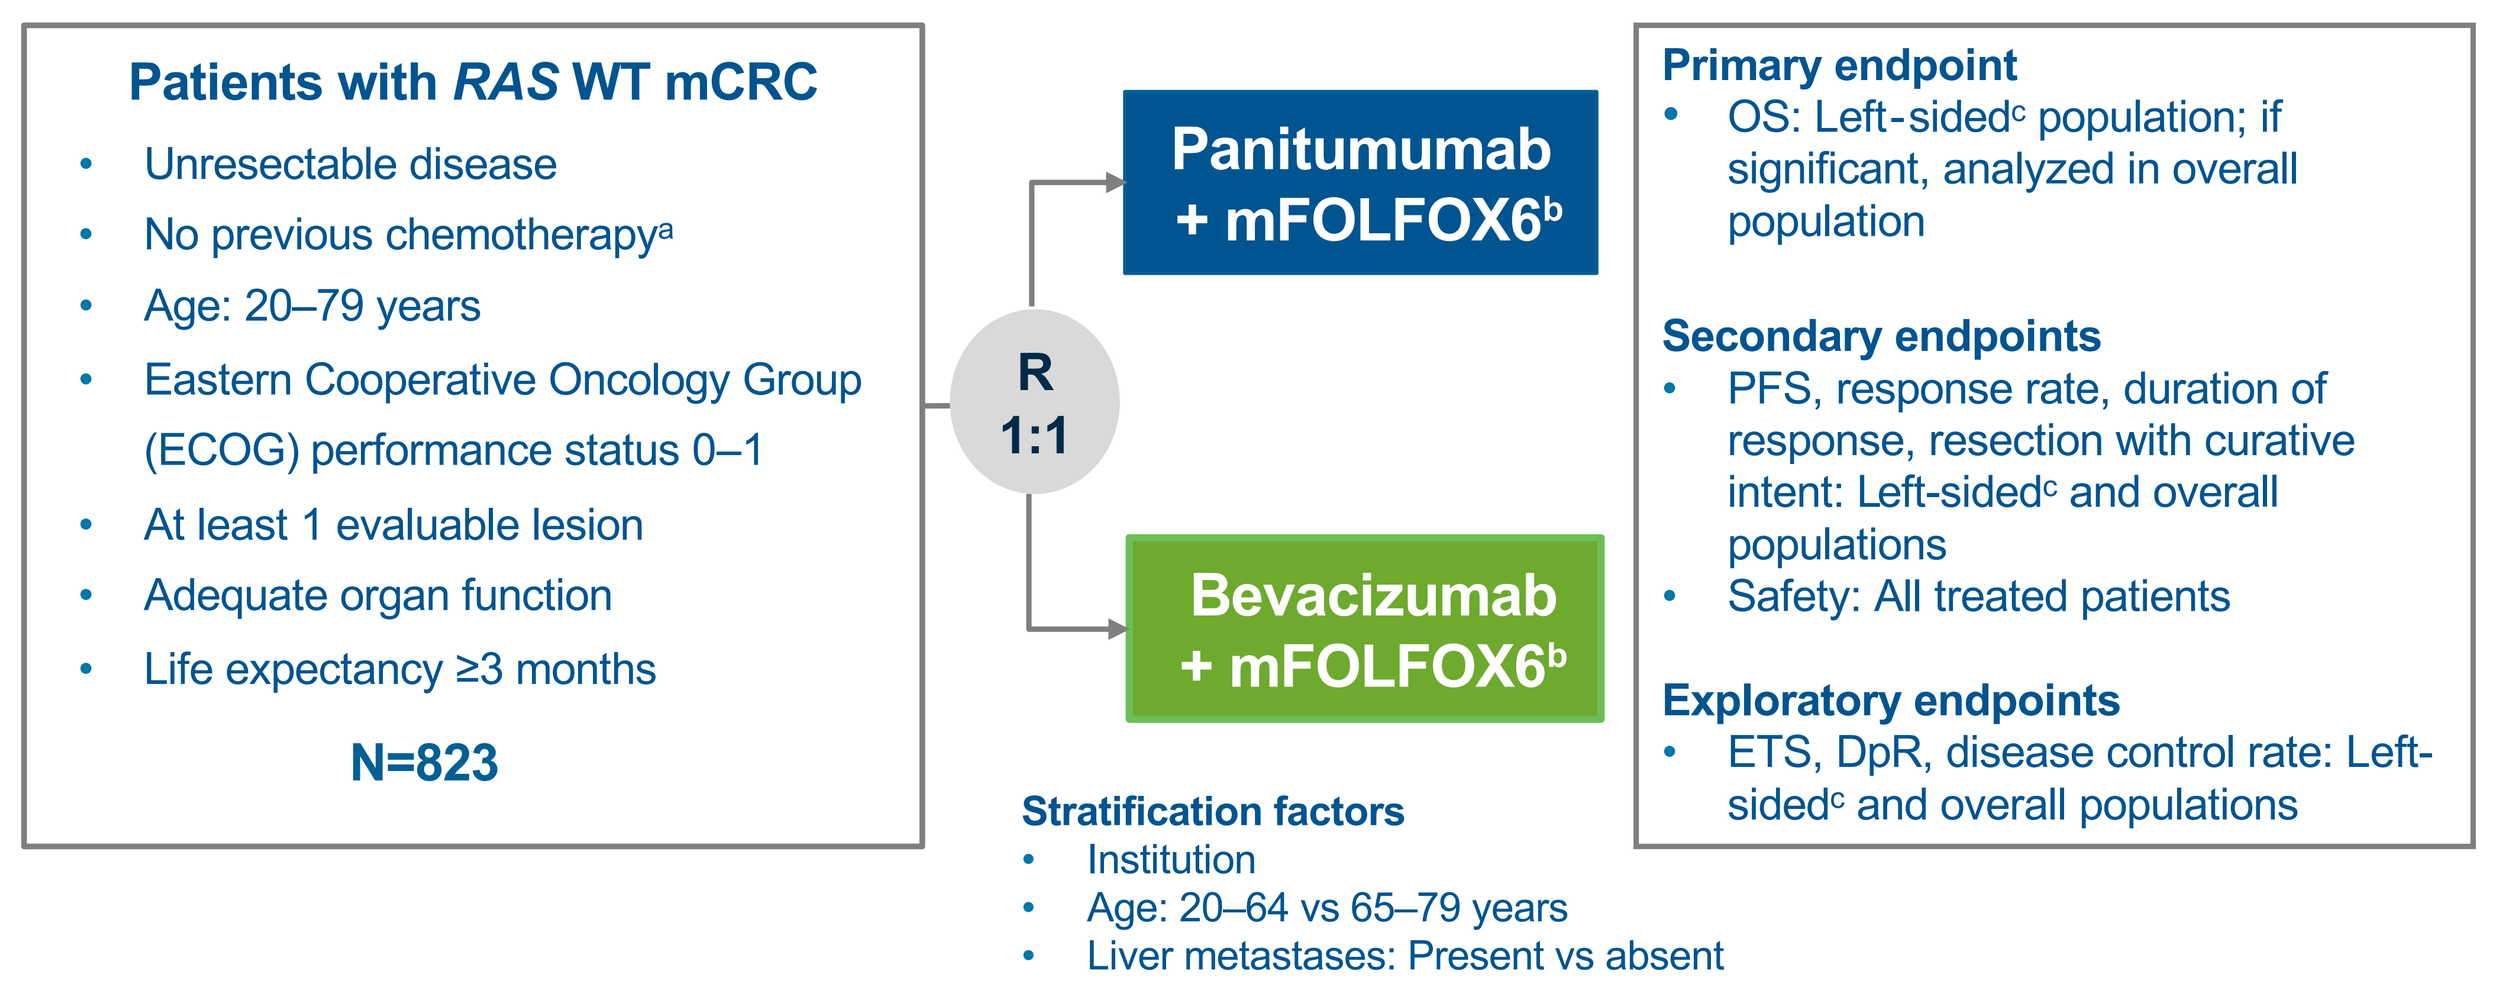

Patients with RAS WT mCRC
Unresectable disease
No previous chemotherapy[a]
Age: 20–79 years
Eastern Cooperative Oncology Group (ECOG) performance status 0–1
At least 1 evaluable lesion
Adequate organ function
Life expectancy ≥3 months
N=823
R 1:1
Panitumumab + mFOLFOX6[b]
Bevacizumab + mFOLFOX6[b]
Stratification factors
Institution
Age: 20–64 vs 65–79 years
Liver metastases: Present vs absent
Primary endpoint
OS: Left-sided[c] population; if significant, analyzed in overall population
Secondary endpoints
PFS, response rate, duration of response, resection with curative intent: Left-sided[c] and overall populations
Safety: All treated patients
Exploratory endpoints
ETS, DpR, disease control rate: Left-sided[c] and overall populations

[a] Adjuvant fluoropyrimidine monotherapy allowed if completed >6 months before enrollment. [b] Until disease progression, unacceptable toxicity, withdrawal of consent or investigator's judgment or curative intent resection. [c] Primary tumor in descending colon, sigmoid colon, rectosigmoid, and rectum.
Data cutoff: January 14, 2022

- The relative change in the sum of the longest diameters of target lesions was analyzed between baseline and 8 weeks of treatment
- ETS was defined as ≥30% decrease in the sum of the longest diameters of target lesions at week 8; OS and PFS were compared in patients with or without ETS with stratification by treatment arm and primary tumor location (left-sided vs right-sided vs overall)
- DpR was defined as the maximum post-baseline percent decrease in the sum diameter of target lesions
- No formal hypothesis testing was performed
- Descriptive *P* values were determined based on a two-sided stratified log-rank test

Results

- In patients with left-sided mCRC, the rate of ETS was higher with panitumumab than with bevacizumab **(Table 2)**
- A similar trend was observed in the overall population

Table 2: Early tumor shrinkage at week 8

Parameter	Left-sided population		Right-sided population		Overall population	
	Panitumumab + mFOLFOX6 (n=312)	Bevacizumab + mFOLFOX6 (n=292)	Panitumumab + mFOLFOX6 (n=84)	Bevacizumab + mFOLFOX6 (n=103)	Panitumumab + mFOLFOX6 (n=400)	Bevacizumab + mFOLFOX6 (n=402)
Patients with ETS, n	201	111	30	42	234	156
Rate of ETS, % (95% CI)	64 (59–70)	38 (32–44)	36 (26–47)	41 (31–51)	59 (54–63)	39 (34–44)

- Demographic and baseline characteristics across treatment arms for ETS evaluable patients are shown in **Table 3**

Table 3: Demographic and baseline characteristics

Characteristic	Left-sided population				Right-sided population				Overall population			
	Panitumumab + mFOLFOX6		Bevacizumab + mFOLFOX6		Panitumumab + mFOLFOX6		Bevacizumab + mFOLFOX6		Panitumumab + mFOLFOX6		Bevacizumab + mFOLFOX6	
	ETS n=201	No ETS n=111	ETS n=111	No ETS n=181	ETS n=30	No ETS n=54	ETS n=42	No ETS n=61	ETS n=234	No ETS n=166	ETS n=156	No ETS n=246
Age category, n (%)												
20–64 years	89 (44.3)	49 (44.1)	41 (36.9)	86 (47.5)	11 (36.7)	15 (27.8)	16 (38.1)	23 (37.7)	100 (42.7)	64 (38.6)	58 (37.2)	110 (44.7)
65–79 years	112 (55.7)	62 (55.9)	70 (63.1)	95 (52.5)	19 (63.3)	39 (72.2)	26 (61.9)	38 (62.3)	134 (57.3)	102 (61.4)	98 (62.8)	136 (55.3)
Female, n (%)	57 (28.4)	47 (42.3)	35 (31.5)	56 (30.9)	13 (43.3)	30 (55.6)	18 (42.9)	24 (39.3)	71 (30.3)	77 (46.4)	54 (34.6)	80 (32.5)
ECOG performance status, n (%)												
0	169 (84.1)	92 (82.9)	91 (82.0)	140 (77.3)	28 (93.3)	37 (68.5)	35 (83.3)	47 (77.0)	199 (85.0)	129 (77.7)	128 (82.1)	191 (77.6)
1	32 (15.9)	19 (17.1)	20 (18.0)	41 (22.7)	2 (6.7)	16 (29.6)	7 (16.7)	14 (23.0)	35 (15.0)	36 (21.7)	28 (17.9)	55 (22.4)
Primary tumor location, n (%)												
Left-sided	202 (100.0)	111 (100.0)	111 (100.0)	181 (100.0)	0	0	0	0	201 (85.9)	111 (66.9)	111 (71.2)	181 (73.6)
Right-sided	0	0	0	0	30 (100.0)	54 (100.0)	42 (100.0)	61 (100.0)	30 (12.8)	54 (32.5)	42 (26.9)	61 (24.8)
Number of metastatic organs, n (%)												
1	98 (48.8)	57 (51.4)	58 (52.3)	89 (49.2)	14 (46.7)	26 (48.1)	20 (47.6)	24 (39.3)	113 (48.3)	83 (50.0)	79 (50.6)	115 (46.7)
≥2	103 (51.2)	54 (48.6)	53 (47.7)	92 (50.8)	16 (53.3)	28 (51.9)	22 (52.4)	37 (60.7)	121 (51.7)	83 (50.0)	77 (49.4)	131 (53.3)
Metastatic site, n (%)												
Liver	164 (81.6)	61 (55.0)	86 (77.5)	120 (66.3)	20 (66.7)	29 (53.7)	26 (61.9)	40 (65.6)	185 (79.1)	90 (54.2)	115 (73.7)	163 (66.3)
Liver as the only metastatic site	71 (35.3)	19 (17.1)	43 (38.7)	46 (25.4)	7 (23.3)	7 (13.0)	7 (16.7)	14 (23.0)	79 (33.8)	26 (15.7)	51 (32.7)	62 (25.2)
Prior treatment, n (%)												
Primary tumor resection	127 (63.2)	58 (52.3)	71 (64.0)	122 (67.4)	19 (63.3)	32 (59.3)	30 (71.4)	43 (70.5)	148 (63.2)	91 (54.8)	104 (66.7)	168 (68.3)
Radiotherapy	1 (0.5)	1 (0.9)	1 (0.9)	2 (1.1)	0	0	0	0	1 (0.4)	1 (0.6)	1 (0.6)	2 (0.8)
Adjuvant chemotherapy	10 (5.0)	7 (6.3)	6 (5.4)	10 (5.5)	2 (6.7)	3 (5.6)	0	3 (4.9)	12 (5.1)	10 (6.0)	7 (4.5)	13 (5.3)

- In the left-sided population, patients with ETS had longer PFS and OS than those without ETS, regardless of treatment with panitumumab or bevacizumab **(Figure 2)**

Figure 2: Survival outcomes by ETS in patients in the left-sided population

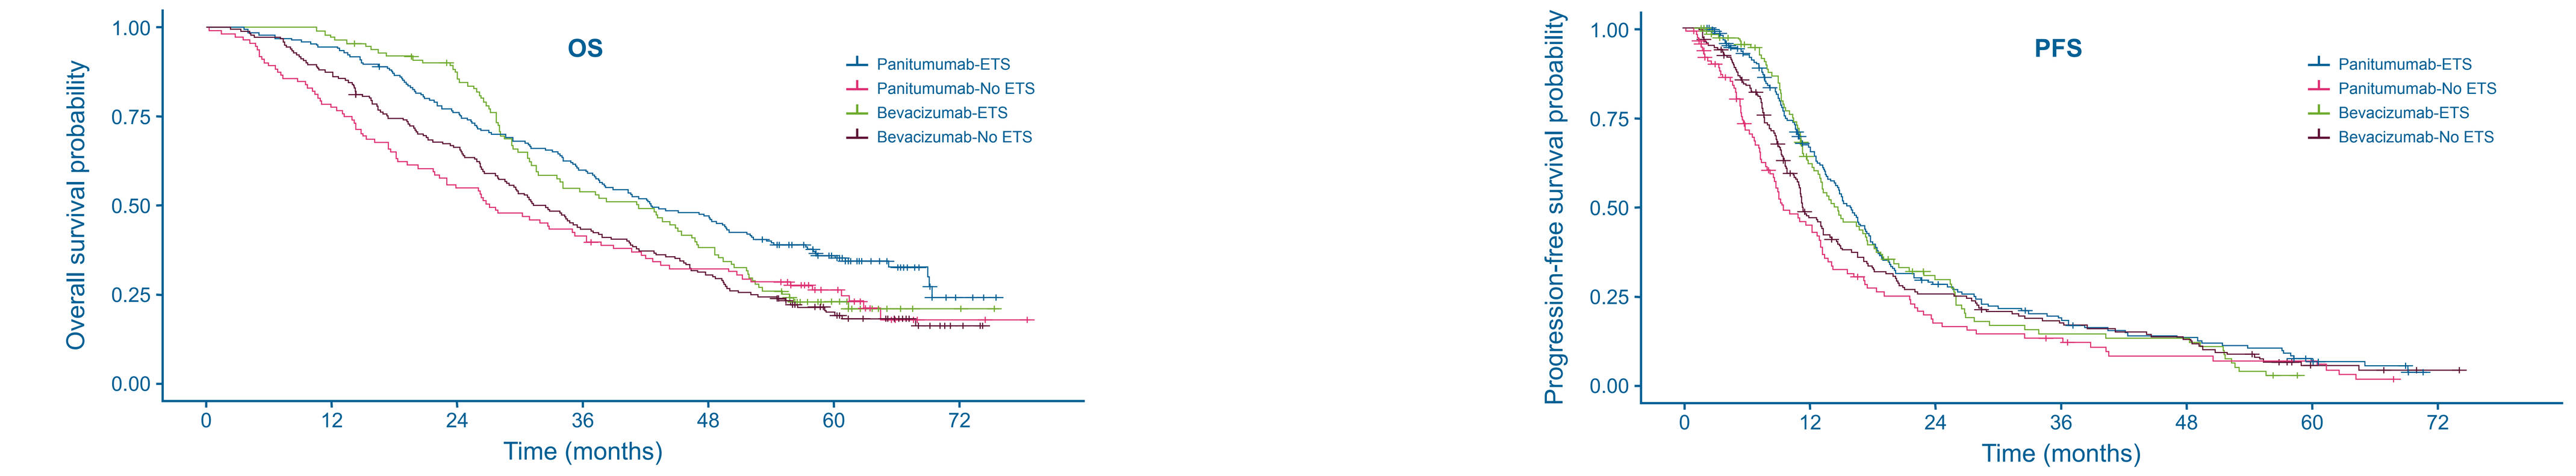

OS
PFS
Overall survival probability
Progression-free survival probability
Time (months)
Panitumumab-ETS
Panitumumab-No ETS
Bevacizumab-ETS
Bevacizumab-No ETS

OS – No. at risk

	0	12	24	36	48	60	72
Panitumumab-ETS	201	190	152	120	94	51	3
Panitumumab-No ETS	111	86	61	46	35	17	2
Bevacizumab-ETS	111	108	93	58	41	14	2
Bevacizumab-No ETS	181	158	119	78	55	26	3

Parameter	Panitumumab + mFOLFOX6 ETS	Panitumumab + mFOLFOX6 No ETS	Bevacizumab + mFOLFOX6 ETS	Bevacizumab + mFOLFOX6 No ETS
Median OS, mo (95% CI)	42.6 (37.5–49.7)	27.1 (21.8–36.3)	41.3 (31.6–46.5)	32.4 (27.9–36.8)
HR (95% CI); *P* value	0.56 (0.42–0.75); <0.001		0.69 (0.52–0.91); 0.010	

PFS – No. at risk

	0	12	24	36	48	60	72
Panitumumab-ETS	201	106	43	27	18	8	0
Panitumumab-No ETS	111	43	16	11	6	5	0
Bevacizumab-ETS	111	61	26	12	11	0	0
Bevacizumab-No ETS	181	78	41	28	20	5	1

Parameter	Panitumumab + mFOLFOX6 ETS	Panitumumab + mFOLFOX6 No ETS	Bevacizumab + mFOLFOX6 ETS	Bevacizumab + mFOLFOX6 No ETS
Median PFS, mo (95% CI)	15.7 (13.9–17.6)	9.6 (8.1–13.1)	14.5 (12.6–17.6)	11.5 (10.9–13.5)
HR (95% CI); *P* value	0.62 (0.48–0.81); <0.001		0.81 (0.62–1.06); 0.094	

- In the right-sided population, patients in the panitumumab arm who had ETS had longer OS than patients without ETS **(Figure 3)**

Figure 3: Survival outcomes by ETS in patients in the right-sided population

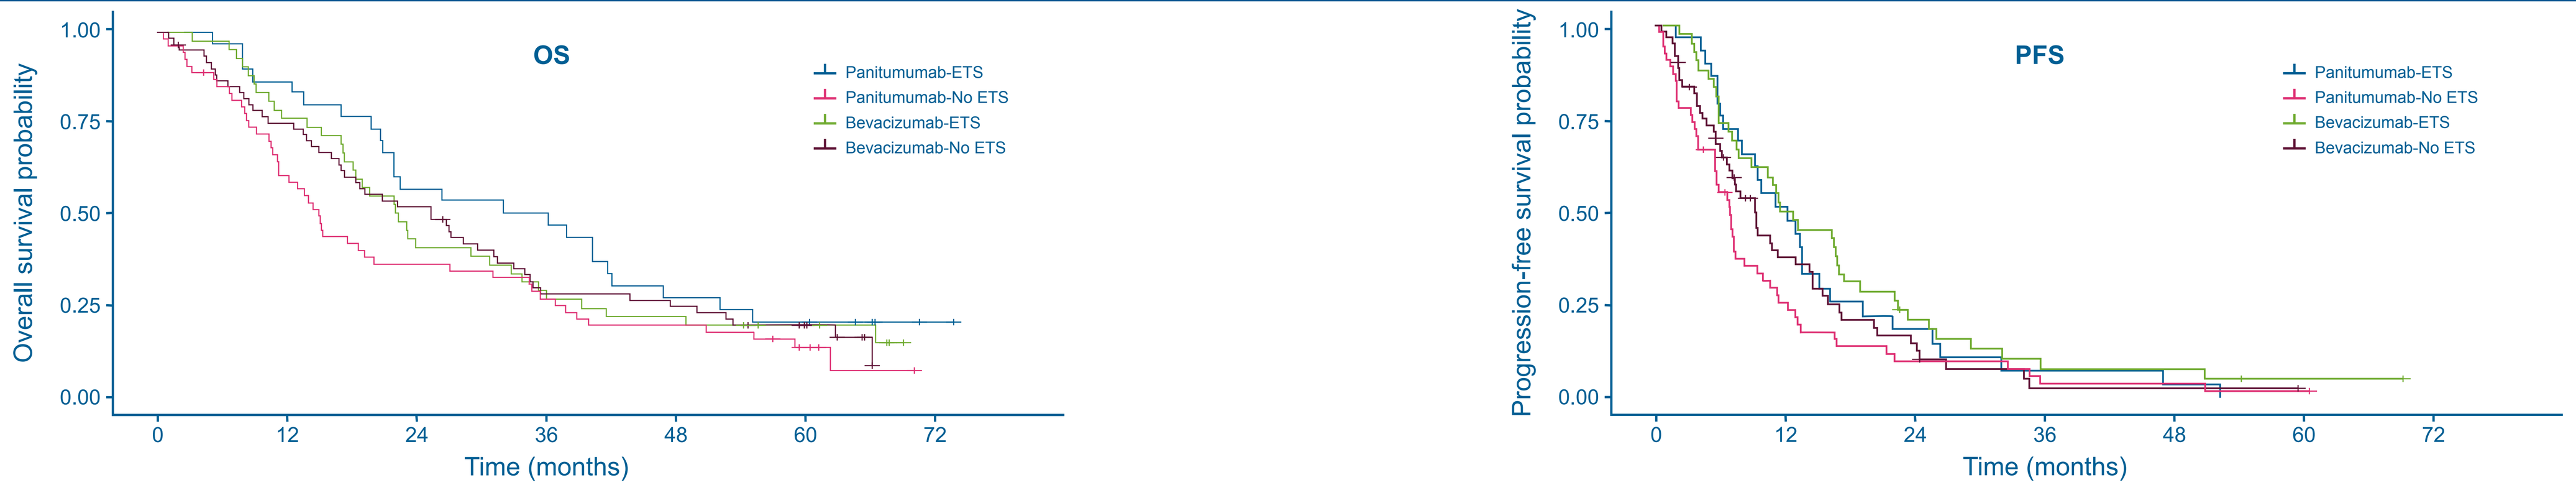

OS
PFS
Overall survival probability
Progression-free survival probability
Time (months)
Panitumumab-ETS
Panitumumab-No ETS
Bevacizumab-ETS
Bevacizumab-No ETS

OS – No. at risk

	0	12	24	36	48	60	72
Panitumumab-ETS	30	26	17	15	8	6	1
Panitumumab-No ETS	54	32	19	14	10	5	0
Bevacizumab-ETS	42	32	18	12	9	6	0
Bevacizumab-No ETS	61	45	31	16	14	8	0

Parameter	Panitumumab + mFOLFOX6 ETS	Panitumumab + mFOLFOX6 No ETS	Bevacizumab + mFOLFOX6 ETS	Bevacizumab + mFOLFOX6 No ETS
Median OS, mo (95% CI)	34.1 (21.0–42.2)	15.2 (11.3–20.2)	22.3 (17.4–30.9)	25.5 (16.9–31.6)
HR (95% CI); *P* value	0.47 (0.28–0.79); 0.005		0.94 (0.60–1.45); 0.746	

PFS – No. at risk

	0	12	24	36	48	60	72
Panitumumab-ETS	30	14	5	2	1	0	0
Panitumumab-No ETS	54	13	5	2	2	1	0
Bevacizumab-ETS	42	21	8	3	3	1	0
Bevacizumab-No ETS	61	19	7	1	1	0	0

Parameter	Panitumumab + mFOLFOX6 ETS	Panitumumab + mFOLFOX6 No ETS	Bevacizumab + mFOLFOX6 ETS	Bevacizumab + mFOLFOX6 No ETS
Median PFS, mo (95% CI)	12.2 (7.7–15.2)	6.8 (5.6–8.2)	12.1 (7.6–16.7)	9.2 (6.6–13.0)
HR (95% CI); *P* value	0.66 (0.41–1.07); 0.084		0.70 (0.46–1.07); 0.084	

- In the overall population, patients with ETS had longer PFS and OS than those without ETS, regardless of treatment with panitumumab or bevacizumab **(Figure 4)**

Figure 4: Survival outcomes by ETS in overall population

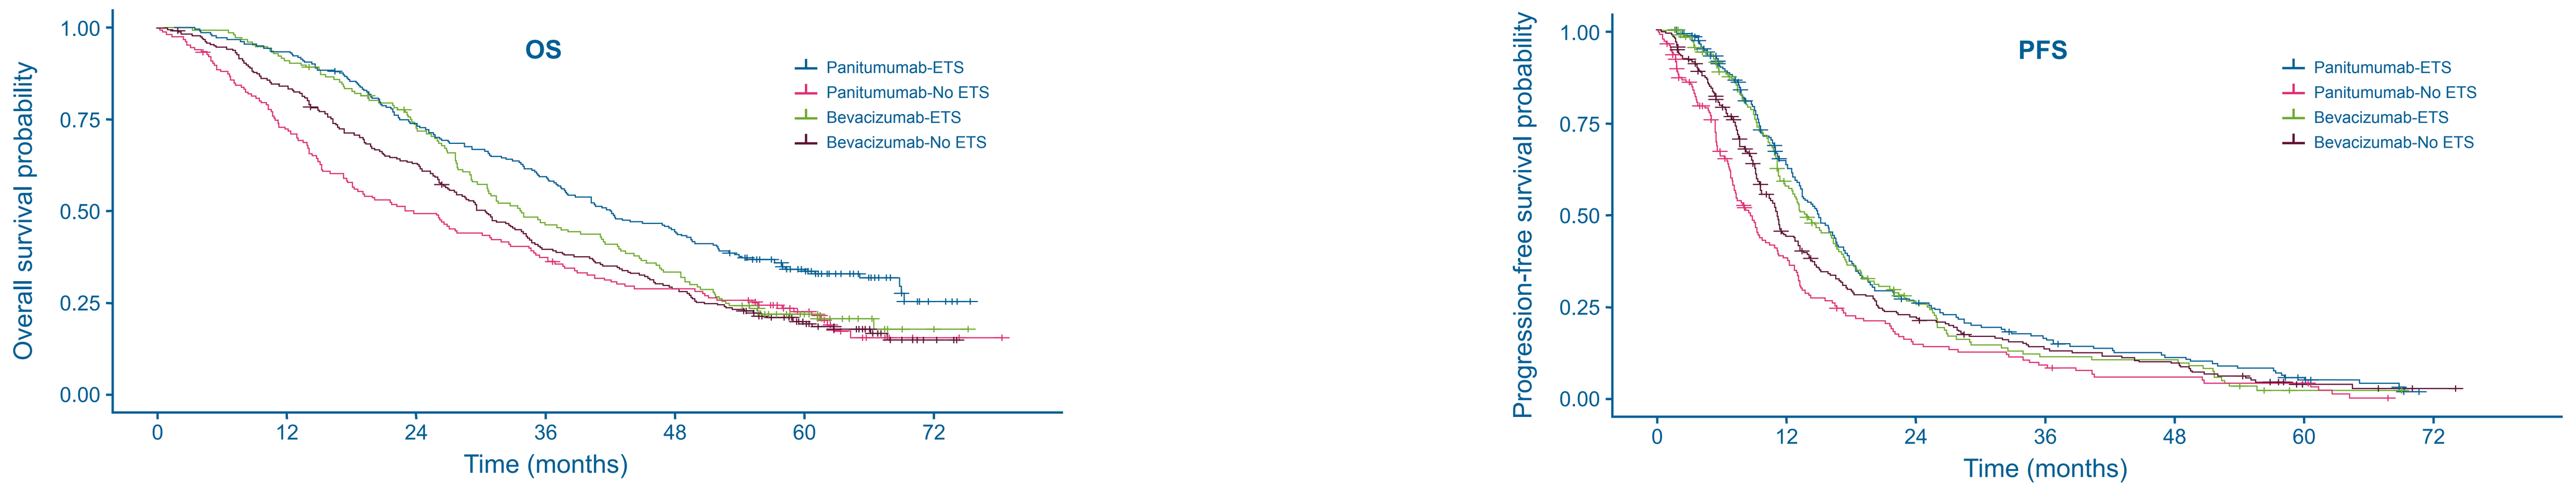

OS
PFS
Overall survival probability
Progression-free survival probability
Time (months)
Panitumumab-ETS
Panitumumab-No ETS
Bevacizumab-ETS
Bevacizumab-No ETS

OS – No. at risk

	0	12	24	36	48	60	72
Panitumumab-ETS	234	219	172	138	104	58	4
Panitumumab-No ETS	166	119	81	61	46	22	2
Bevacizumab-ETS	156	142	112	71	50	20	2
Bevacizumab-No ETS	246	206	153	95	69	34	3

Parameter	Panitumumab + mFOLFOX6 ETS	Panitumumab + mFOLFOX6 No ETS	Bevacizumab + mFOLFOX6 ETS	Bevacizumab + mFOLFOX6 No ETS
Median OS, mo (95% CI)	42.1 (37.5–48.4)	23.1 (17.7–31.0)	33.8 (29.3–41.3)	30.5 (26.9–34.0)
HR (95% CI); *P* value	0.50 (0.39–0.64); <0.001		0.78 (0.62–0.99); 0.036	

PFS – No. at risk

	0	12	24	36	48	60	72
Panitumumab-ETS	234	122	49	30	20	9	0
Panitumumab-No ETS	166	57	22	13	8	6	0
Bevacizumab-ETS	156	83	34	15	14	1	0
Bevacizumab-No ETS	246	99	49	30	21	5	1

Parameter	Panitumumab + mFOLFOX6 ETS	Panitumumab + mFOLFOX6 No ETS	Bevacizumab + mFOLFOX6 ETS	Bevacizumab + mFOLFOX6 No ETS
Median PFS, mo (95% CI)	15.2 (13.6–16.7)	8.9 (7.2–10.6)	14.1 (12.0–16.7)	11.2 (9.8–13.0)
HR (95% CI); *P* value	0.58 (0.47–0.73); <0.001		0.82 (0.66–1.02); 0.055	

- In left-sided and overall populations, DpR was greater with panitumumab compared with bevacizumab **(Figure 5)**
- Among patients with right-sided mCRC, DpR was similar across treatment groups

Figure 5: DpR in mCRC populations

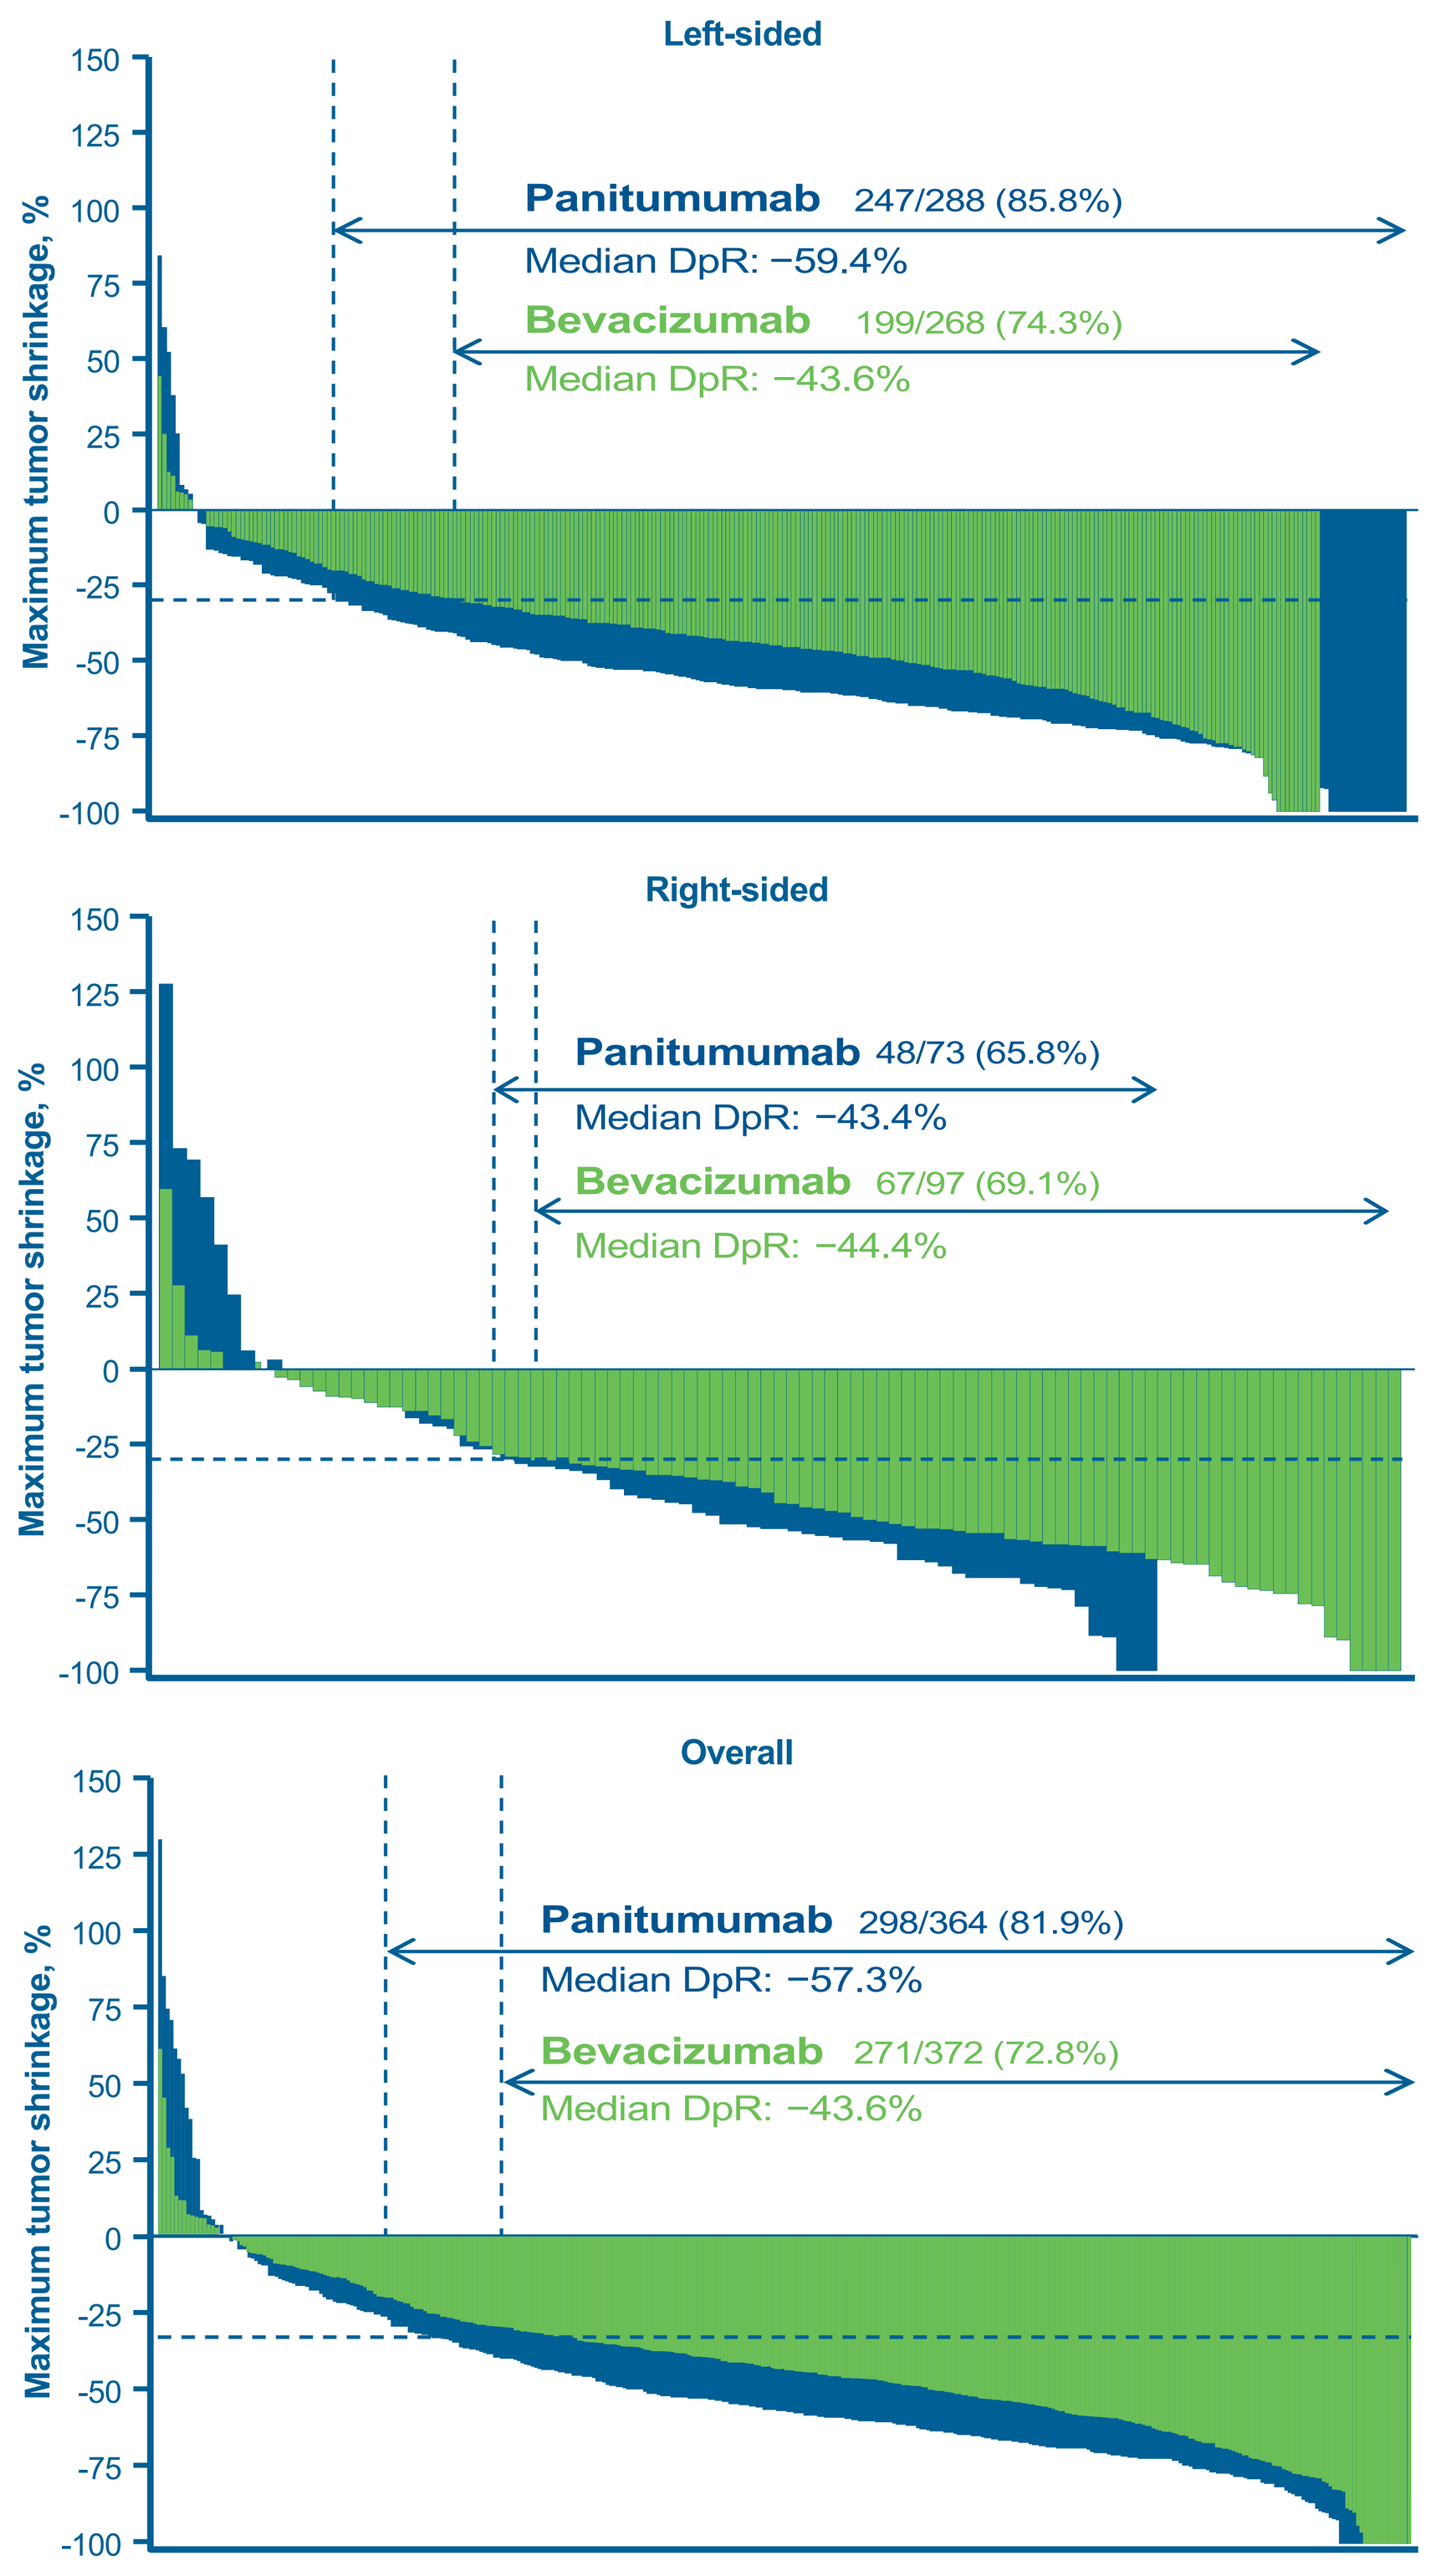

Left-sided
Panitumumab 247/288 (85.8%)
Median DpR: −59.4%
Bevacizumab 199/268 (74.3%)
Median DpR: −43.6%
Right-sided
Panitumumab 48/73 (65.8%)
Median DpR: −43.4%
Bevacizumab 67/97 (69.1%)
Median DpR: −44.4%
Overall
Panitumumab 298/364 (81.9%)
Median DpR: −57.3%
Bevacizumab 271/372 (72.8%)
Median DpR: −43.6%
Maximum tumor shrinkage, %

Horizontal dotted line at 30% indicates response per RECIST v1.1.
Depth of response was assessed in patients with measurable lesions at baseline.

Conclusions

- Regardless of treatment with panitumumab or bevacizumab, patients with ETS had favorable survival outcomes compared with those without ETS
- Patients with left-sided mCRC treated with panitumumab had a higher rate of ETS and a greater DpR compared with patients treated with bevacizumab
- In patients with right-sided mCRC, ETS appeared to predict better survival outcomes with panitumumab but not with bevacizumab
 – This finding highlights the need for additional analyses to identify biomarkers that may predict a benefit with panitumumab in this population

Future Directions: Biomarker Multi-omics Analysis

- A large-scale biomarker analysis is currently underway using plasma and tumor tissue samples collected pre- and post-treatments (NCT02394834)
- Potential biomarkers on outcomes will be reported in upcoming meetings
- Biomarkers that may predict a benefit with panitumumab in patients with mCRC will be identified

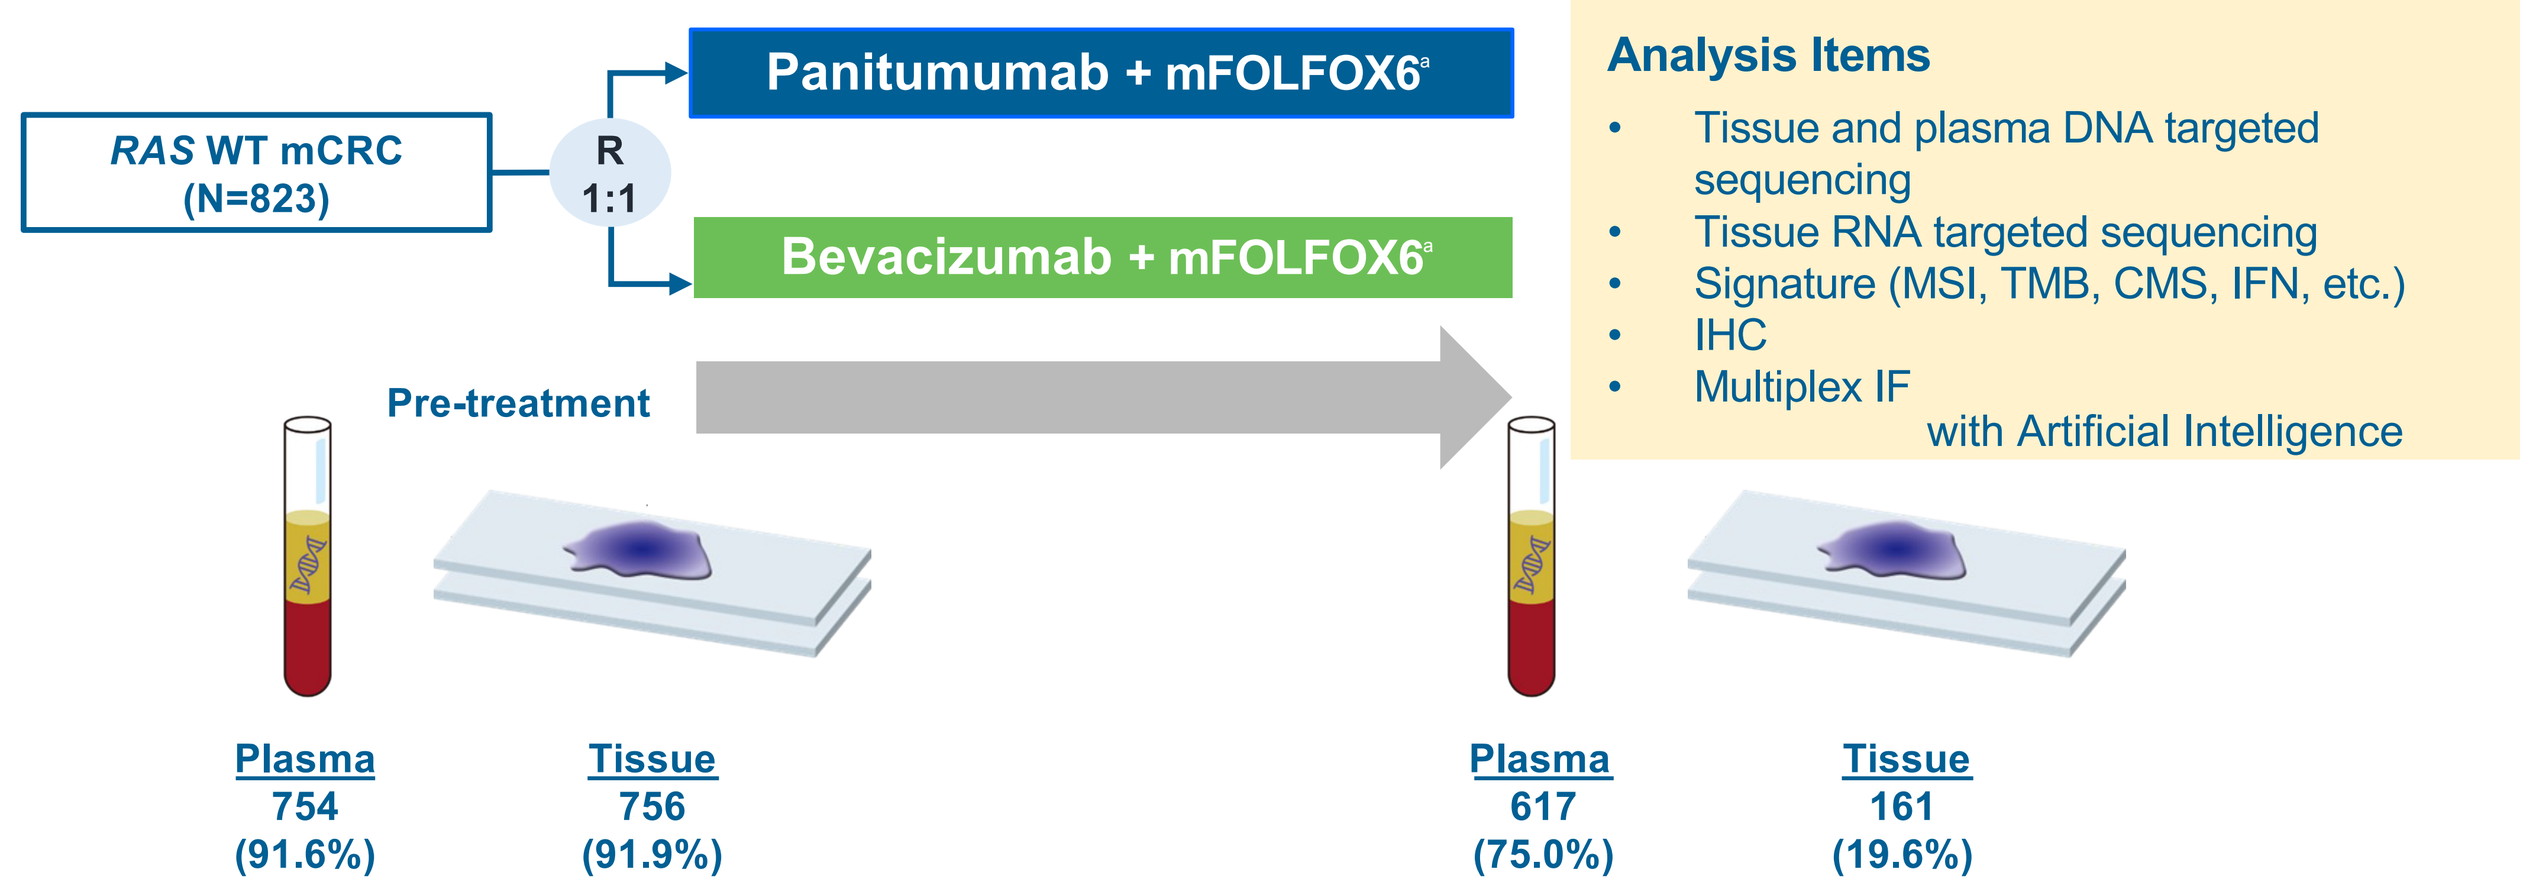

RAS WT mCRC (N=823)
R 1:1
Panitumumab + mFOLFOX6[a]
Bevacizumab + mFOLFOX6[a]
Pre-treatment
Plasma 754 (91.6%)
Tissue 756 (91.9%)
Plasma 617 (75.0%)
Tissue 161 (19.6%)
Analysis Items
Tissue and plasma DNA targeted sequencing
Tissue RNA targeted sequencing
Signature (MSI, TMB, CMS, IFN, etc.)
IHC
Multiplex IF with Artificial Intelligence

[a] Until disease progression, unacceptable toxicity, withdrawal of consent or investigator's judgement or curative intent resection. CMS, consensus molecular subtypes; DNA, deoxyribonucleic acid; IF, immunofluorescence; IFN, interferon gene signature; IHC, immunohistochemistry; mCRC, metastatic colorectal cancer; MSI, microsatellite instability; RNA, ribonucleic acid; TMB, tumor mutational burden; WT, wild type

References

1. Yoshino T, et al. J Clin Oncol. 2022;40(17 suppl):LBA1.
2. Giessen C, et al. Cancer Sci. 2013;104:718-724.
3. Cremolini C, et al. Ann Oncol. 2015;26:1188-1194.

Acknowledgments

The authors would like to thank the patients, their families, and their caregivers; the PARADIGM investigators and their team members at each study site; and colleagues from Takeda. Professional medical writing assistance was provided by Kalpana Vijayan, PhD, and Lauren Gallagher, RPh, PhD, of Peloton Advantage, LLC, an OPEN Health company, Parsippany, NJ, USA, and funded by Millennium Pharmaceuticals, Inc., Cambridge, MA, USA, a wholly owned subsidiary of Takeda Pharmaceutical Company Limited. The study was sponsored by Takeda.

Abbreviations

CI, confidence interval; DpR, depth of response; ECOG, Eastern Cooperative Oncology Group; ETS, Early tumor shrinkage; HR, harzard ratio; mCRC, metastatic colorectal cancer; mFOLFOX6, modified FOLFOX6; NA, not applicable; OS, overall survival; PFS, progression-free survival; WT, wild type

Disclosures

KM: Consulting/advisory role (Chugai Pharma, Astra Zeneca, Ono Pharmaceutical, Amgen); honoraria (Chugai Pharma, Ono Pharmaceutical, Takeda, Lilly, Bayer, Sanofi, Bristol Myers Squibb, Taiho Pharmaceutical); research grant (Taiho Pharmaceutical, Astellas Pharma, Amgen Astellas Biopharma, [rest to institution:] MSD, Daiichi Sankyo, Shionogi, Kyowa Kirin, Gilead Sciences, Merck Serono, Pfizer, Sanofi, PAREXEL, Mediscience Planning, Sumitomo Dainippon Pharma, Solasia Pharma); **JW:** Speakers bureau (Covidien Japan, Johnson & Johnson/Janssen, Lilly Japan); research funding (to institution: Medtronic, TERUMO); **KS:** Honoraria (Bristol Myers Squibb, Takeda); consulting/advisory role (Lilly, Bristol Myers Squibb, Takeda, Pfizer, Ono Pharmaceutical, MSD, Taiho, Novartis, Abbvie, GSK, Daiichi Sankyo, Boehringer Ingelheim, Janssen); research funding (all to institution: MSD, Daiichi Sankyo, Taiho, Chugai Pharma, Ono Pharmaceutical, Astellas Pharma, Medi Science, Eisai, Amgen); **KY:** Honoraria (Chugai Pharma, Daiichi Sankyo, Yakult Honsha, Takeda, Bayer, Merck Serono, Taiho Pharmaceutical, Lilly, Sanofi, Ono Pharmaceutical, MSD, Bristol Myers Squibb); research funding (to institution: Taiho Pharmaceutical); **HO:** The author has no relationships to disclose; **MS:** The author has no relationships to disclose; **HY:** The author has no relationships to disclose; **EO:** Speakers bureau (Chugai Pharma, Lilly Japan, Takeda, Ono Pharmaceutical, Bayer Yakuhin, Bristol Myers Squibb Japan); **TS:** Consulting/advisory role (Takeda); speakers bureau (Chugai Pharma, Lilly Japan, Taiho Oncology, Takeda, Bayer Yakuhin, Ono Yakuhin, Daiichi Sankyo/UCB Japan); **TN:** Honoraria (Chugai Pharma, Taiho Pharmaceutical, Kaken, Daiichi Sankyo, Eli Lilly Japan, Takeda, Merck, Bayer, Boehringer Ingelheim); research funding (to institution: Chugai, Taiho, Kaken, Daiichi-Sankyo, Eli Lilly Japan); **YK:** Speakers bureau (Ono, Taiho, Chugai, Eli Lilly, Bayer Yakuhin); research funding (Ono, Taiho, Daiichi Sankyo, Chugai, IQVIA); **TK:** Honoraria (Chugai, Yakult Honsha, Ono Pharmaceutical, Takeda, Lilly Japan, Taiho Pharmaceutical, Asahi Kasei); research funding (Chugai); **MH:** Employment (Takeda); **JS:** Employment (Takeda); **KY:** Honoraria (Chugai, J-Pharma, Johokiko, Triceps, CMIC Holdings); research funding (Taiho, Boehringer Ingelheim, Takeda, Daiichi Sankyo, Astellas); **KA:** The author has no relationships to disclose; **AO:** The author has no relationships to disclose; **HU:** Speakers bureau (Takeda, Chigai, Taiho); **KT:** Honoraria (Chugai Pharma, Novartis, Takeda, Miyarisan Pharmaceutical, Bristol Myers Squibb Japan, AstraZeneca); **TY:** Honoraria (Chugai Pharma, Merck, Bayer Yakuhin, Ono Pharmaceutical, MSD); research funding (to institution: MSD, Daiichi Sankyo, Ono Pharmaceutical, Taiho Pharmaceutical, Amgen, Sanofi, Pfizer, Genomedia Inc., Sysmex, Nippon Boehringer Ingelheim, Chugai Pharma)

For an e-Print, scan this QR code.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the authors of this poster.

Presented at the 2022 Annual Meeting of the European Society of Medical Oncology (ESMO), 9–13 September, Paris, France

Email for questions or comments: Kei Muro, kmuro@aichi-cc.jp